Results from Phase 2 Study of BPM31510 in Combination With Standard of Care (SOC) RT and TMZ in Newly Diagnosed Glioblastoma (GB)

727TiP

Brian Stocksdale[1], Adam Cohen[2], Andrew Brenner[6], Chirag G. Patil[3], Brian Vaillant[4], Kevin Elmore[5], Rupa Juthani[7], David Reardon[8], Ryan Kendle[9], Alexander Spira[10], Jedrzej Wykretowicz[11], Poornima Tekumalla[13], Greg Miller[13], Michael A. Kiebish[13], Valentina Zhukova-Harrill[12], Bing Nie[13], Megan K. Cox[13], Viatcheslav R. Akmaev[13], Niven R. Narain[13,14,15], Brian Berman[13], Seema Nagpal[1]

[1]Division of Neuro-Oncology, Department of Neurology and Neurological Sciences, Stanford University, Stanford, CA; [2]Inova Schar Cancer Institute, Fairfax, VA; [3]Cedars-Sinai Medical Center, Los Angeles, CA; [4]Texas Oncology-South Central, Austin, TX; [5]Icahn School of Medicine at Mt Sinai, New York, NY; [6]University of Texas Health Science Center at San Antonio, Mays Cancer Center at UT Health San Antonio MD Anderson, San Antonio, TX; [7]Valley Health Hospital, Ridgewood, NJ; [8]Dana Farber Cancer Institute, Boston, MA; [9]Sansum Clinic, Santa Barbara, CA; [10]Virginia Cancer Specialists, Fairfax, VA; [11]Virginia Oncology Associates, Norfolk, VA; [12]Aixial Group, Greensboro, NC; [13]BPGbio Inc., Waltham, MA; [14]Department of Molecular Biology, University of Miami, Miller School of Medicine, Miami, FL; [15]Department of Dermatology and Cutaneous Surgery, University of Miami, Miller School of Medicine, Miami, FL

Low Expression of CoQ10 Genes in GB Correlates with Poor Survival

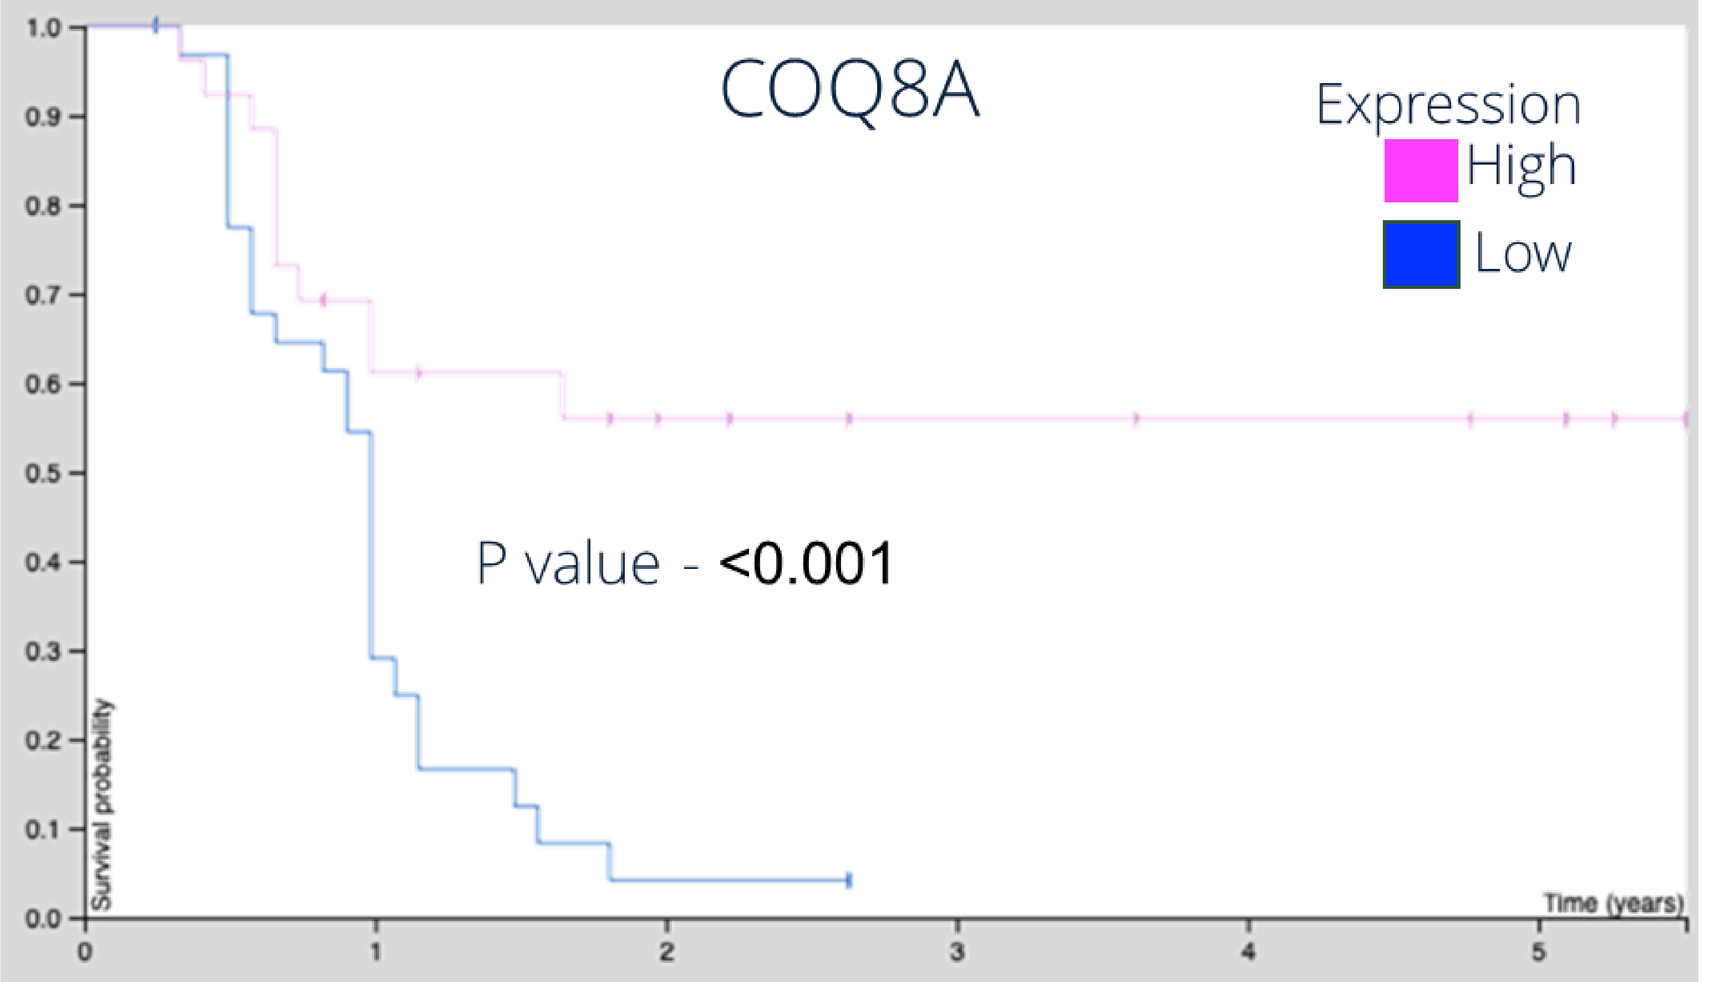
COQ8A
Expression
High
Low
Survival probability
P value - <0.001
Time (years)

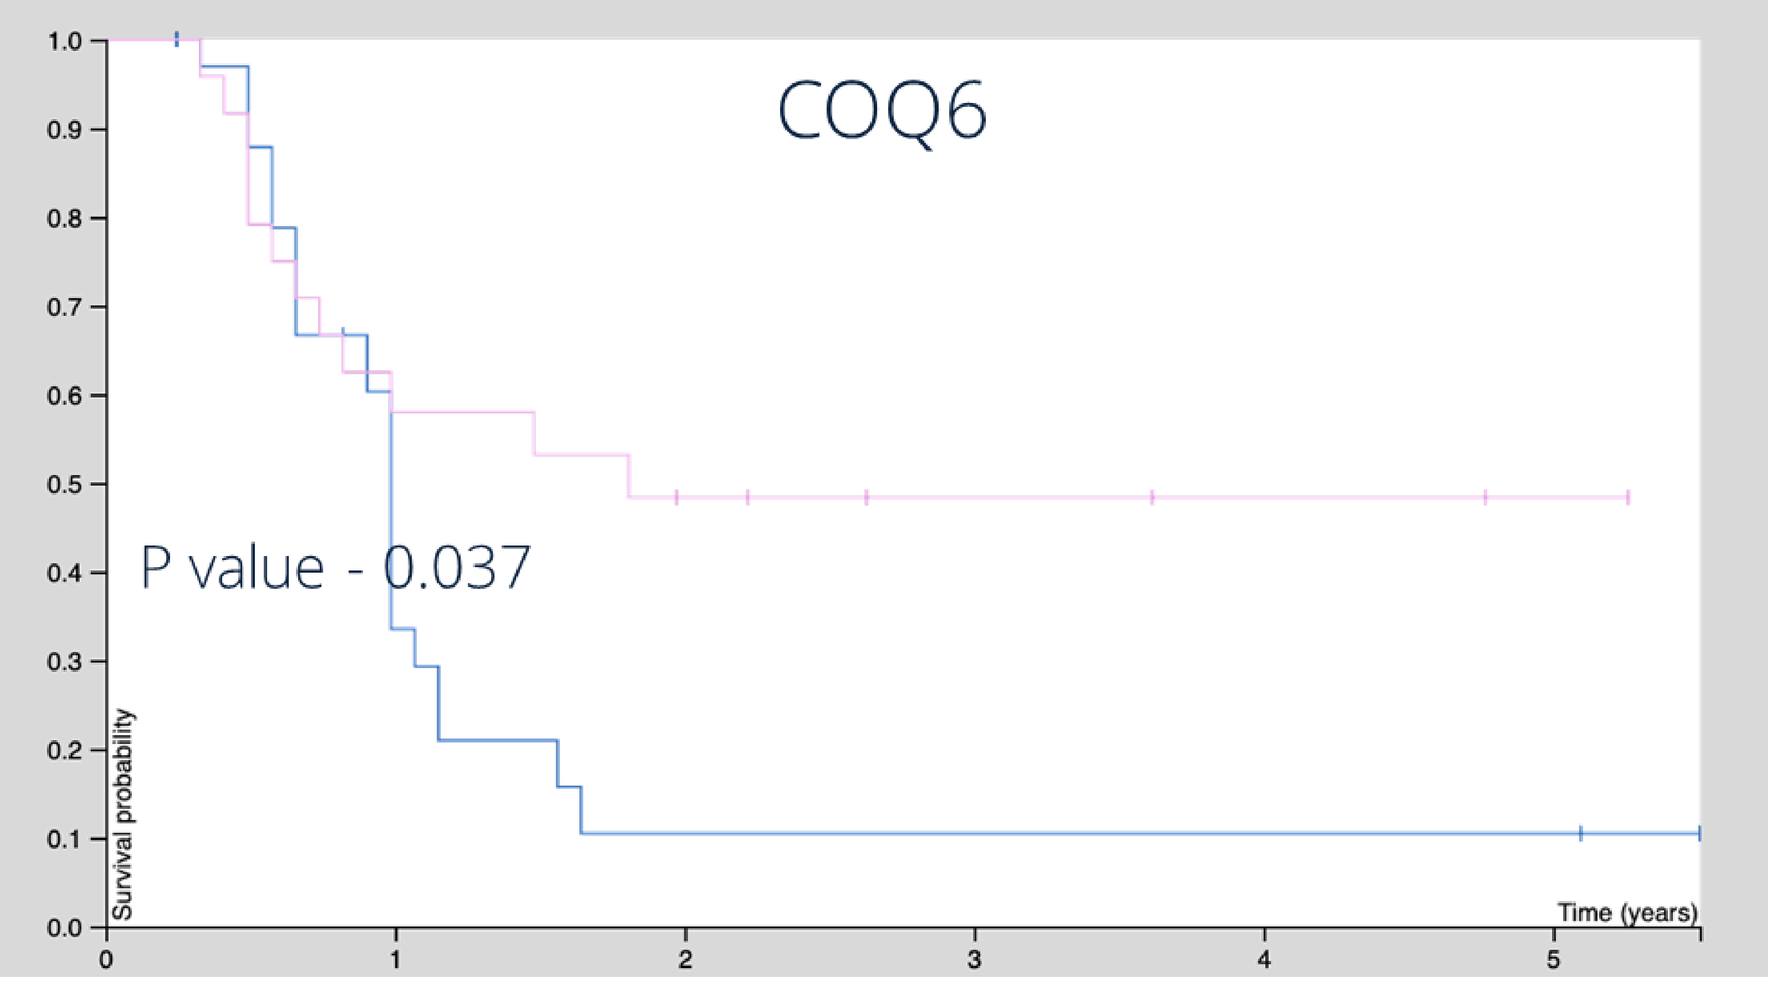
COQ6
P value - 0.037
Time (years)

Human Proteome Atlas TCGA analysis COQ10 synthesis enzymes mRNA expression level and survival.

BPM31510 Mechanism of Action: Reversing the Warburg Effect

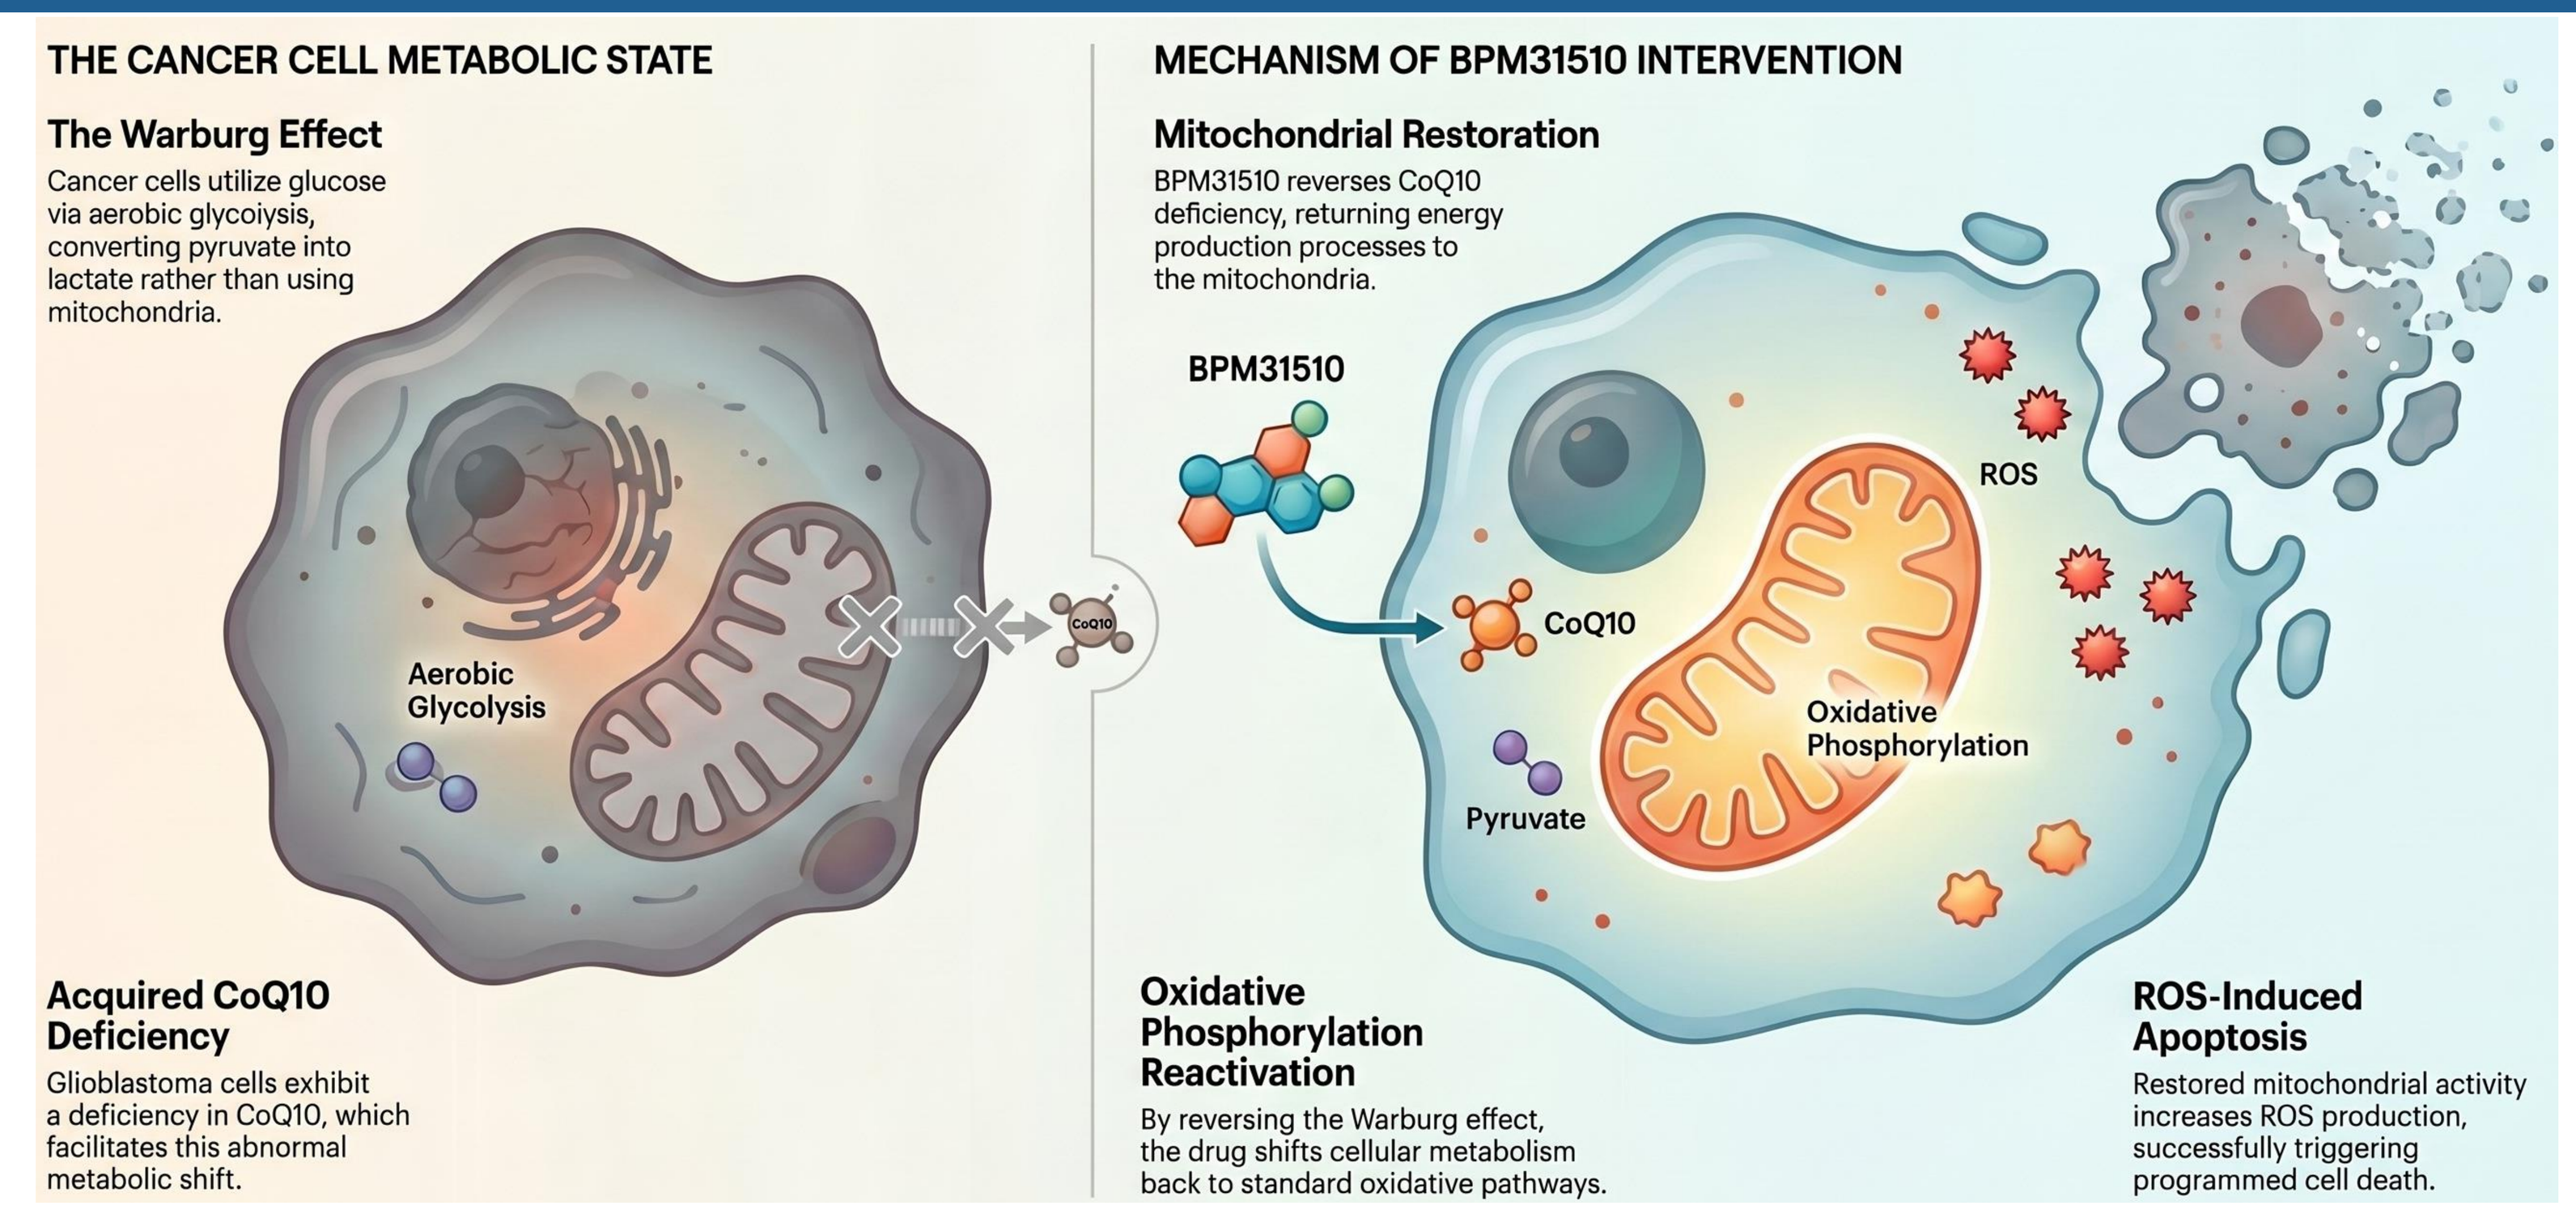
THE CANCER CELL METABOLIC STATE
The Warburg Effect
Cancer cells utilize glucose via aerobic glycoiysis, converting pyruvate into lactate rather than using mitochondria.
Aerobic Glycolysis
Acquired CoQ10 Deficiency
Glioblastoma cells exhibit a deficiency in CoQ10, which facilitates this abnormal metabolic shift.
MECHANISM OF BPM31510 INTERVENTION
Mitochondrial Restoration
BPM31510 reverses CoQ10 deficiency, returning energy production processes to the mitochondria.
BPM31510
CoQ10
ROS
Pyruvate
Oxidative Phosphorylation
Oxidative Phosphorylation Reactivation
By reversing the Warburg effect, the drug shifts cellular metabolism back to standard oxidative pathways.
ROS-Induced Apoptosis
Restored mitochondrial activity increases ROS production, successfully triggering programmed cell death.

BPM31510IV-11 Study

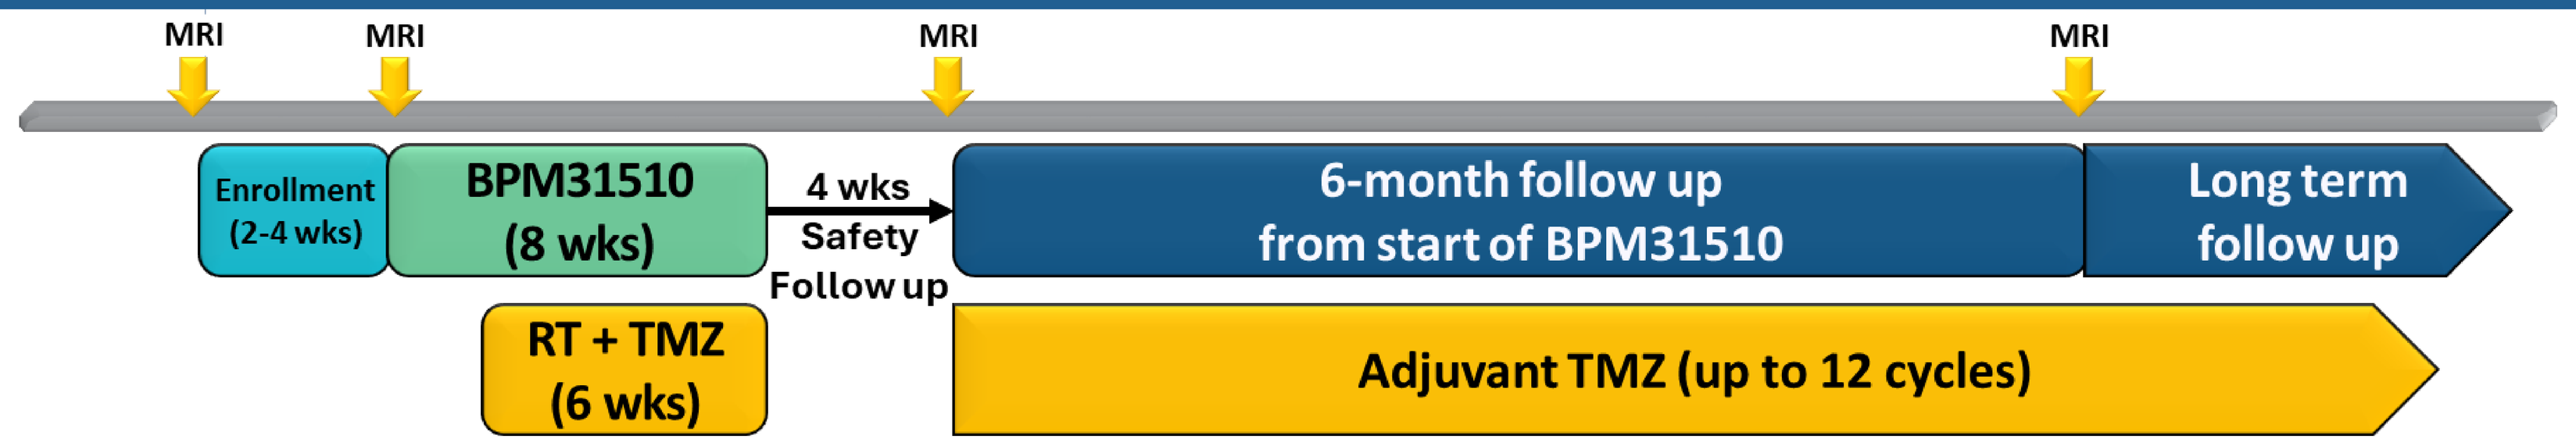
MRI
MRI
MRI
MRI
Enrollment (2-4 wks)
BPM31510 (8 wks)
4 wks Safety Follow up
6-month follow up from start of BPM31510
Long term follow up
RT + TMZ (6 wks)
Adjuvant TMZ (up to 12 cycles)

Abbreviations: MRI = magnetic resonance imaging; RT = radiation therapy; TMZ = temozolomide; PFS = progression free survival

Study Design:
- An open-label study to assess efficacy of BPM31510 plus standard of care RT and TMZ in newly diagnosed GB.
- The study enrolled 51 subjects for ~90% power in rejecting the null hypothesis of PFS6 of ≤ 30%.

Primary Endpoint:
- PFS at 6 and 12 months (defined as meeting RANO 1.0 criteria for complete response, partial response, or stable disease).

Secondary Endpoints:
- Overall survival (defined as time from start of BPM31510 to date of death or last follow-up).
- Safety and tolerability of BPM31510 + vitamin K1 administered with standard RT and TMZ in newly diagnosed GB.

Demographics : ITT Population (N=51)

MGMT Status	Methylated	Unmethylated	Unknown	Total
Age at screening				
N	17	33	1	51
Mean (SD)	65 (13.8)	58.2 (9.36)	64.0 (-)	60.6 (11.29)
Median	70.0	58.0	64.0	64.0
Sex at birth – n (%)				
Female	8 (47.1)	14 (42.4)	1 (100)	23 (45.1)
Male	9 (52.9)	19 (57.6)	0	28 (54.9)
Race – n (%)				
American Indian or Alaska Native	1 (5.9)	0	0	1 (2.0)
Asian	0	2 (6.1)	0	2 (3.9)
Black or African American	0	0	0	0
White	14 (82.4)	27 (81.8)	1 (100)	42 (82.4)
Others	0	4 (12.1)	0	4 (7.8)
Not Reported	2 (11.8)	0	0	2 (3.9)

Tumor Response at 6 Months Based on RANO 1.0 - PP Population

MGMT Status	Methylated	Unmethylated	Total
N	15	28	44
Best overall response - n (%)			
Complete response (CR)	0	1 (3.6)	1 (2.3)
Partial response (PR)	0	3 (10.7)	3 (6.8)
Stable disease (SD)	5 (33.3)	10 (35.7)	15 (34.1)
Progressive disease (PD)	9 (60.0)	13 (46.4)	22 (50.0)
Not evaluable (NE)	1 (6.7)	0	1 (2.3)
Overall response rate (CR or PR) - n (%)	0	4 (14.3)	4 (9.1)
95 % CI for ORR	(0.0, 21.8)	(4.0, 32.7)	(2.5, 21.7)
Disease control rate (CR or PR or SD) - n (%)	5 (33.3)	14 (50.0)	19 (43.2)
95 % CI for DCR	(11.8, 61.6)	(30.6, 69.4)	(28.3, 59.0)

PP = Per-Protocol (subjects completed first 2 weeks BPM31510 and started RT and TMZ); CI = Confidence Interval; 95% CI is estimated using the Clopper-Pearson method. Overall Response is assessed up to day 183 after first BPM31510 infusion; Patients with no post-baseline tumor assessment are included in the population denominator but are not shown in any response categories; therefore, the sum of response categories may not equal the total population.

Tumor Response at 12 Months Based on RANO 1.0 - PP Population

MGMT Status:	Methylated	Unmethylated	Total
N	15	28	44
Best overall response - n (%)			
Complete response (CR)	0	1 (3.6)	1 (2.3)
Partial response (PR)	0	4 (14.3)	4 (9.1)
Stable disease (SD)	5 (33.3)	12 (42.9)	17 (38.6)
Progressive disease (PD)	9 (60.0)	10 (35.7)	19 (43.2)
Not evaluable (NE)	1 (6.7)	0	1 (2.3)
Overall response rate (CR or PR) - n (%)	0	5 (17.9)	5 (11.4)
95 % CI for ORR	(0.0, 21.8)	(6.1, 36.9)	(3.8, 24.6)
Disease control rate (CR or PR or SD) - n (%)	5 (33.3)	17 (60.7)	22 (50.0)
95 % CI for DCR	(11.8, 61.6)	(40.6, 78.5)	(34.6, 65.4)

PP = Per-Protocol (subjects completed first 2 weeks BPM31510 and started RT and TMZ); CI = Confidence Interval; 95% CI is estimated using the Clopper-Pearson method. Overall Response is assessed up to day 365 after first BPM31510 infusion; Patients with no post-baseline tumor assessment are included in the population denominator but are not shown in any response categories; therefore, the sum of response categories may not equal the total population.

Safety: ITT Population (N=51)

Number of patients with:	Overall N=51; n (%)	Related to BPM31510; n (%)
Any TEAE	50 (98.0)	37 (72.5)
Any serious TEAE	18 (35.3)	7 (13.7)
Any TEAE leading to treatment discontinuation	5 (9.8)	3 (5.9)
Any TEAE leading to death	4 (7.8)	1 (2.0)*
Any AESIs	5 (9.8)	

TEAE by NCI-CTCAE	grade 1	grade 2	grade 3	grade 4	grade 5
	48 (94.1)	33 (64.7)	24 (47.1)	6 (11.8)	4 (7.8)

TEAE by System Preferred Term	Any CTC grade; n (%) [Events]	TEAESI by Preferred Term	Any CTC grade; n (%) [Events]
Fatigue	21 (41.2) [26]	Platelet count decreased	10 (19.6) [19]
Nausea	20 (39.2) [23]	Alanine aminotransferase increased	6 (11.8) [14]
Headache	18 (35.3) [22]	Lymphocyte count decreased	6 (11.8) [22]
Constipation	18 (35.3) [20]	Aspartate aminotransferase increased	5 (9.8) [7]
Alopecia	11 (21.6) [12]	Activated partial thromboplastin time prolonged	3 (5.9) [4]
Seizure	8 (15.7) [9]		
Pruritus	7 (13.7) [10]	International normalized ratio increased	3 (5.9) [3]
Brain oedema	7 (13.7) [7]	Haemorrhage intracranial	1 (2) [5]

Note: MedDRA v29.0. CTCAE v5.0, TEAE = Treatment-Emergent Adverse Event, If there is more than one TEAE with maximum NCI CTCAE grade within a system organ class (SOC), the patient is counted only once under that SOC. If there is more than one TEAE with maximum NCI CTCAE grade within a SOC and preferred term (PT), the patient is counted only once in that SOC and PT. Only PTs > 9% are included in table. [Events] presents the number of TEAEs by primary SOC and PT regardless of severity. *Patient expired due to progressive disease with pancytopenia "possibly related" to BPM31510.

Overall Survival PP Population (N=44)

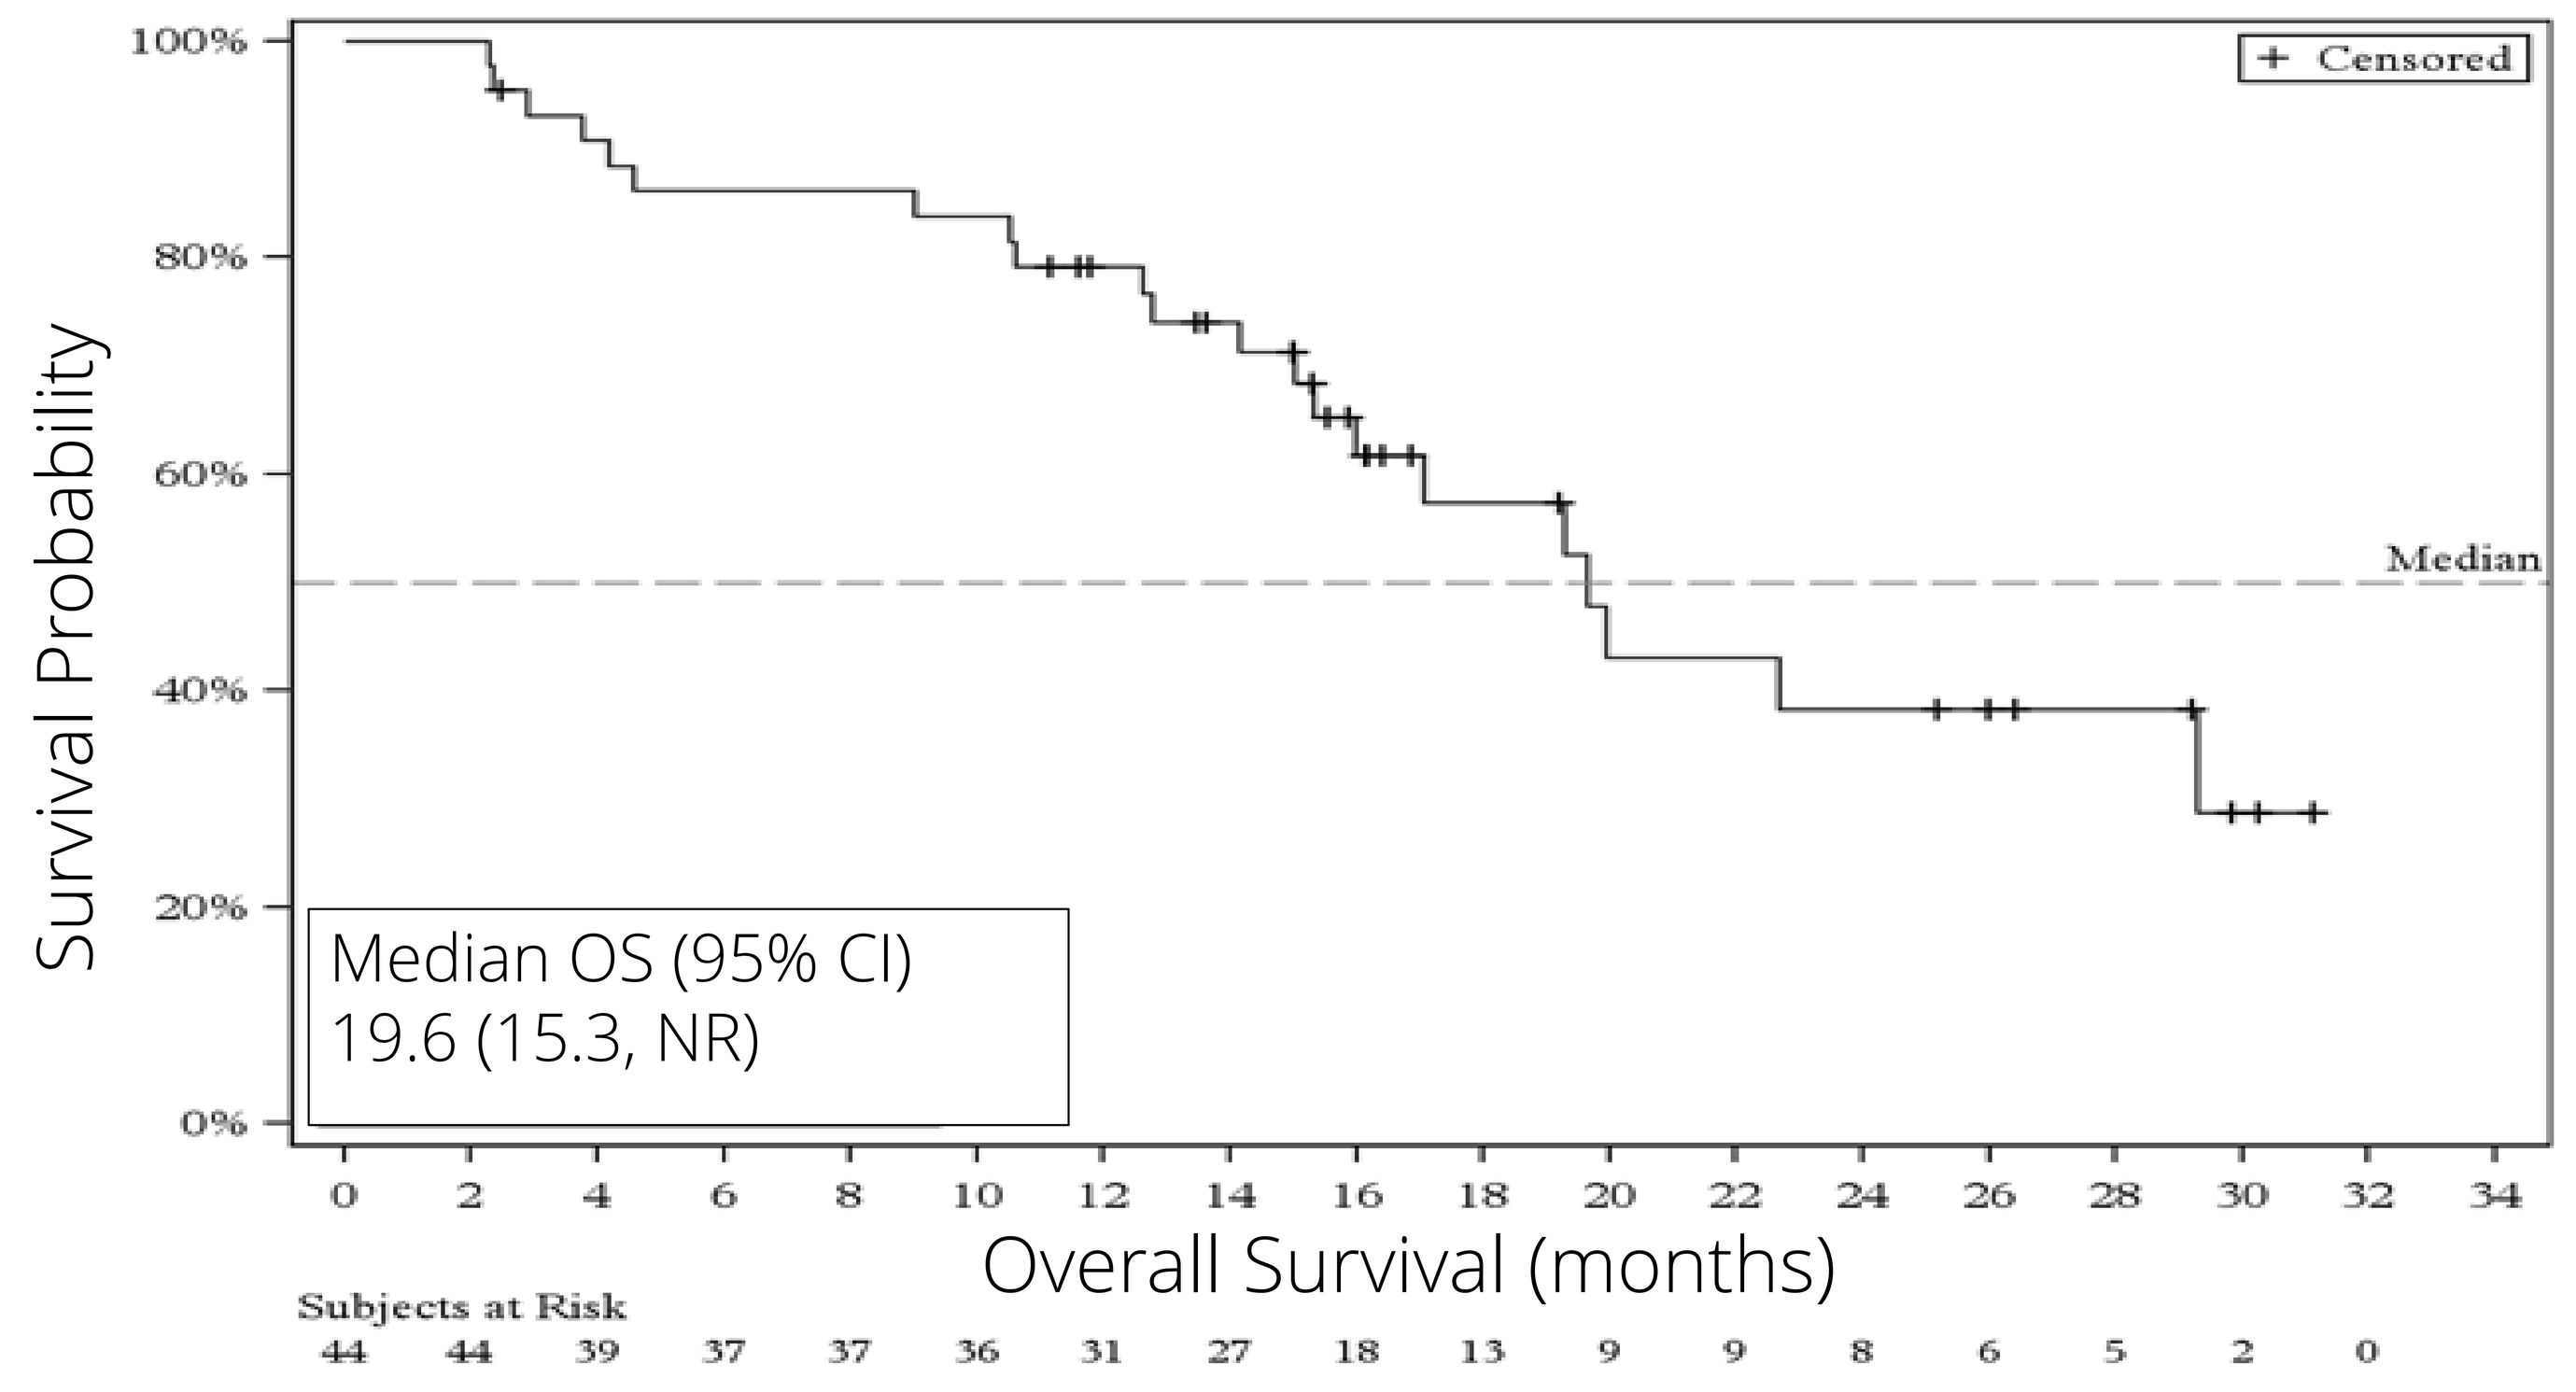
+ Censored
Median
Survival Probability
Median OS (95% CI)
19.6 (15.3, NR)
Overall Survival (months)
Subjects at Risk
44 44 39 37 37 36 31 27 18 13 9 9 8 6 5 2 0

PP= Per Protocol (subjects completed at least 2 weeks BPM31510 and started RT and TMZ), CI = Confidence Interval, OS = Overall Survival, NR = Not Reached; OS is the time from first date of BPM31510 exposure to death

Overall Survival PP Population MGMT: Methylated (N=15) vs Unmethylated (N=28)

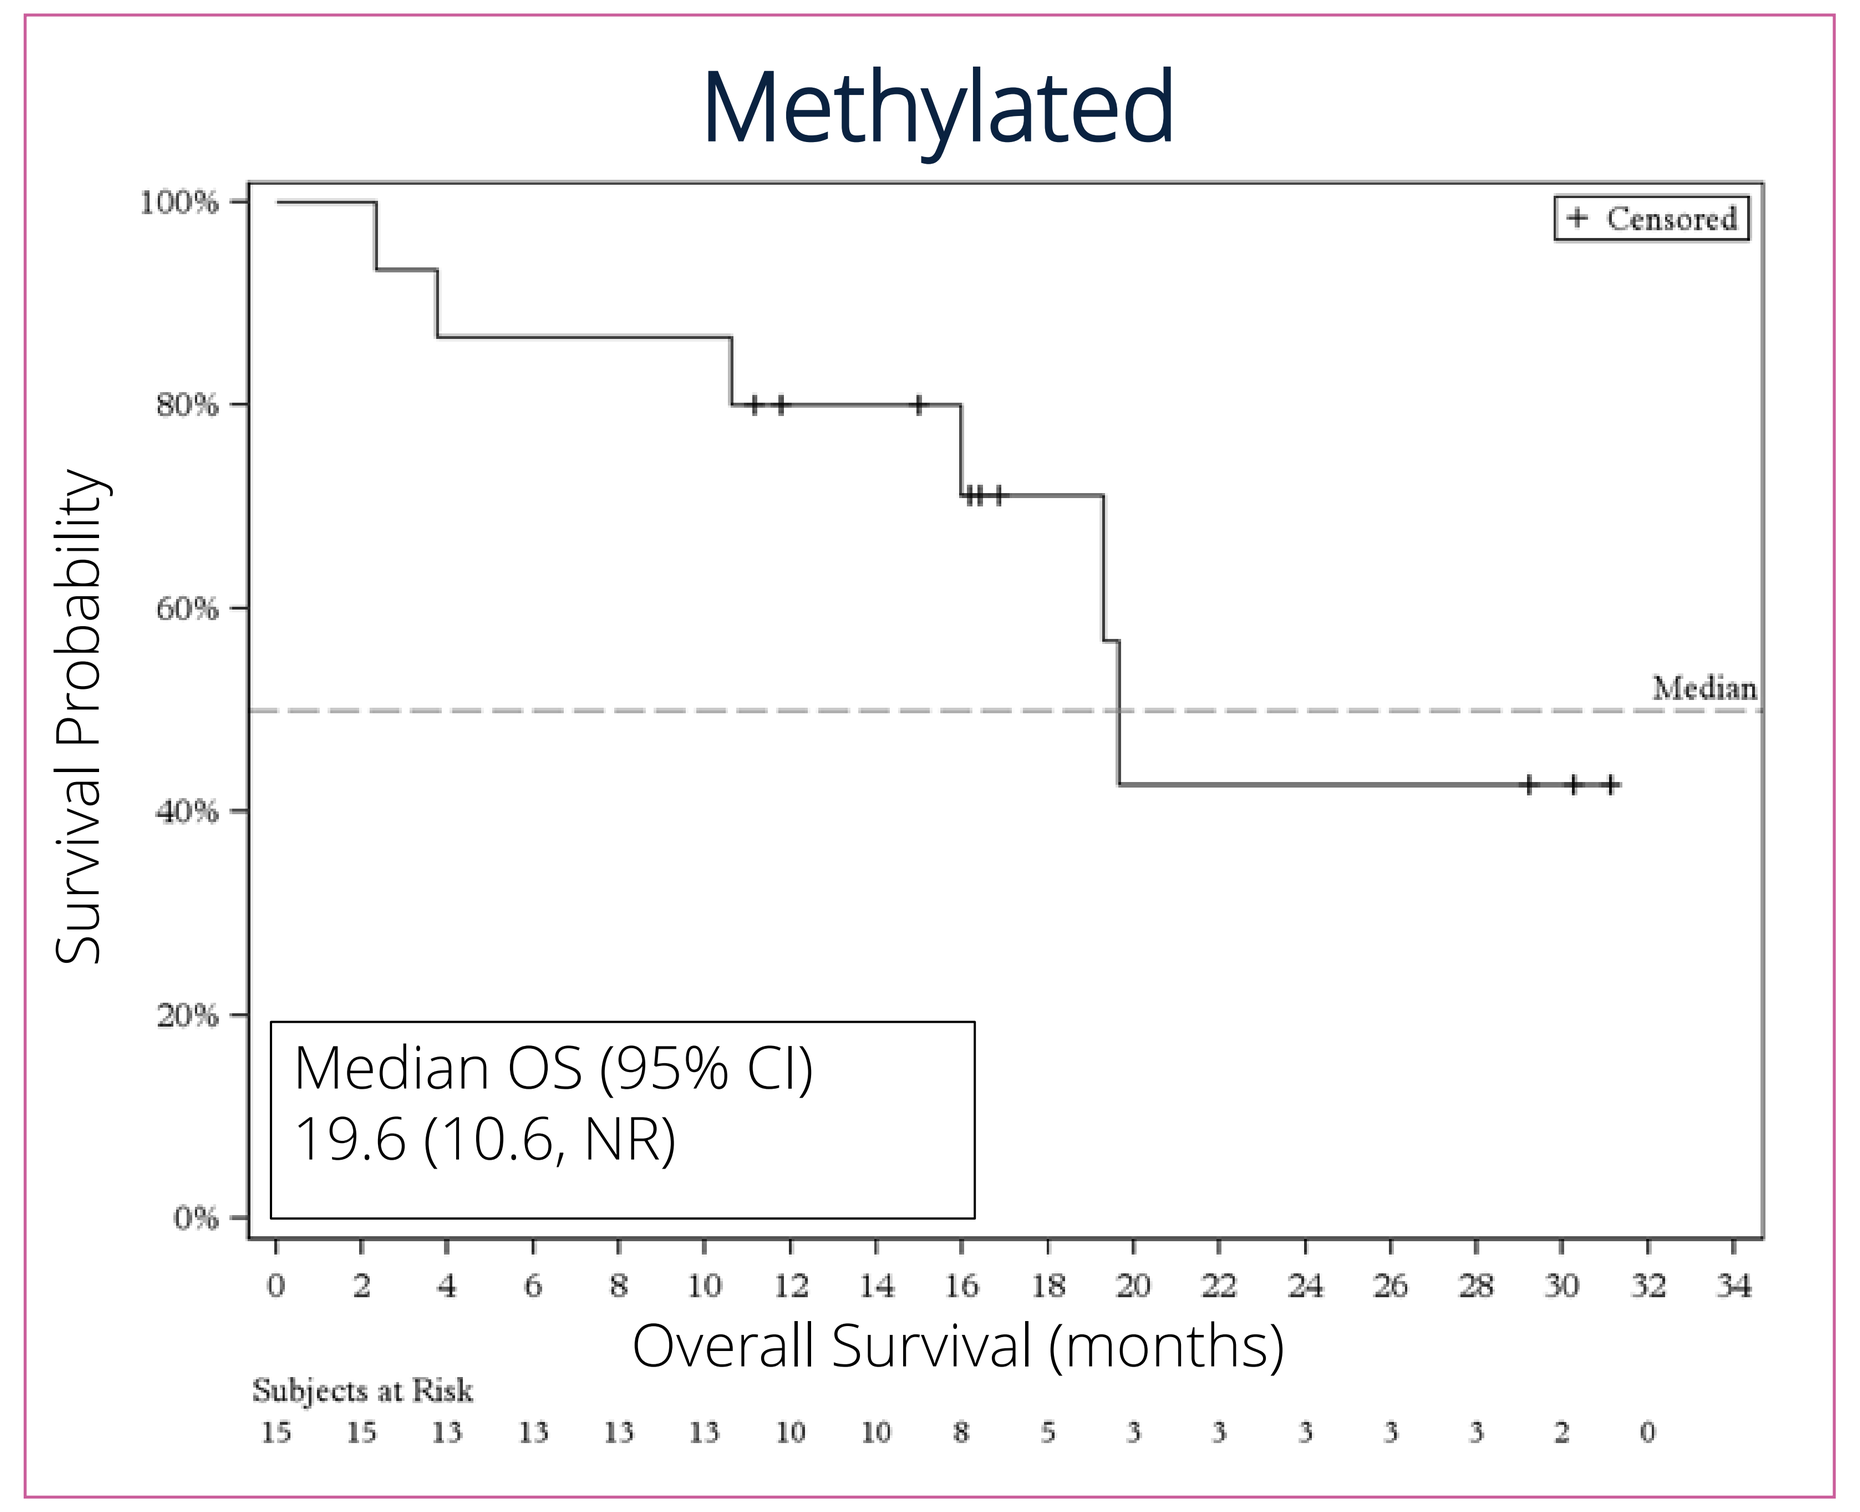
Methylated
+ Censored
Median
Survival Probability
Median OS (95% CI)
19.6 (10.6, NR)
Overall Survival (months)
Subjects at Risk
15 15 13 13 13 13 10 10 8 5 3 3 3 3 3 2 0

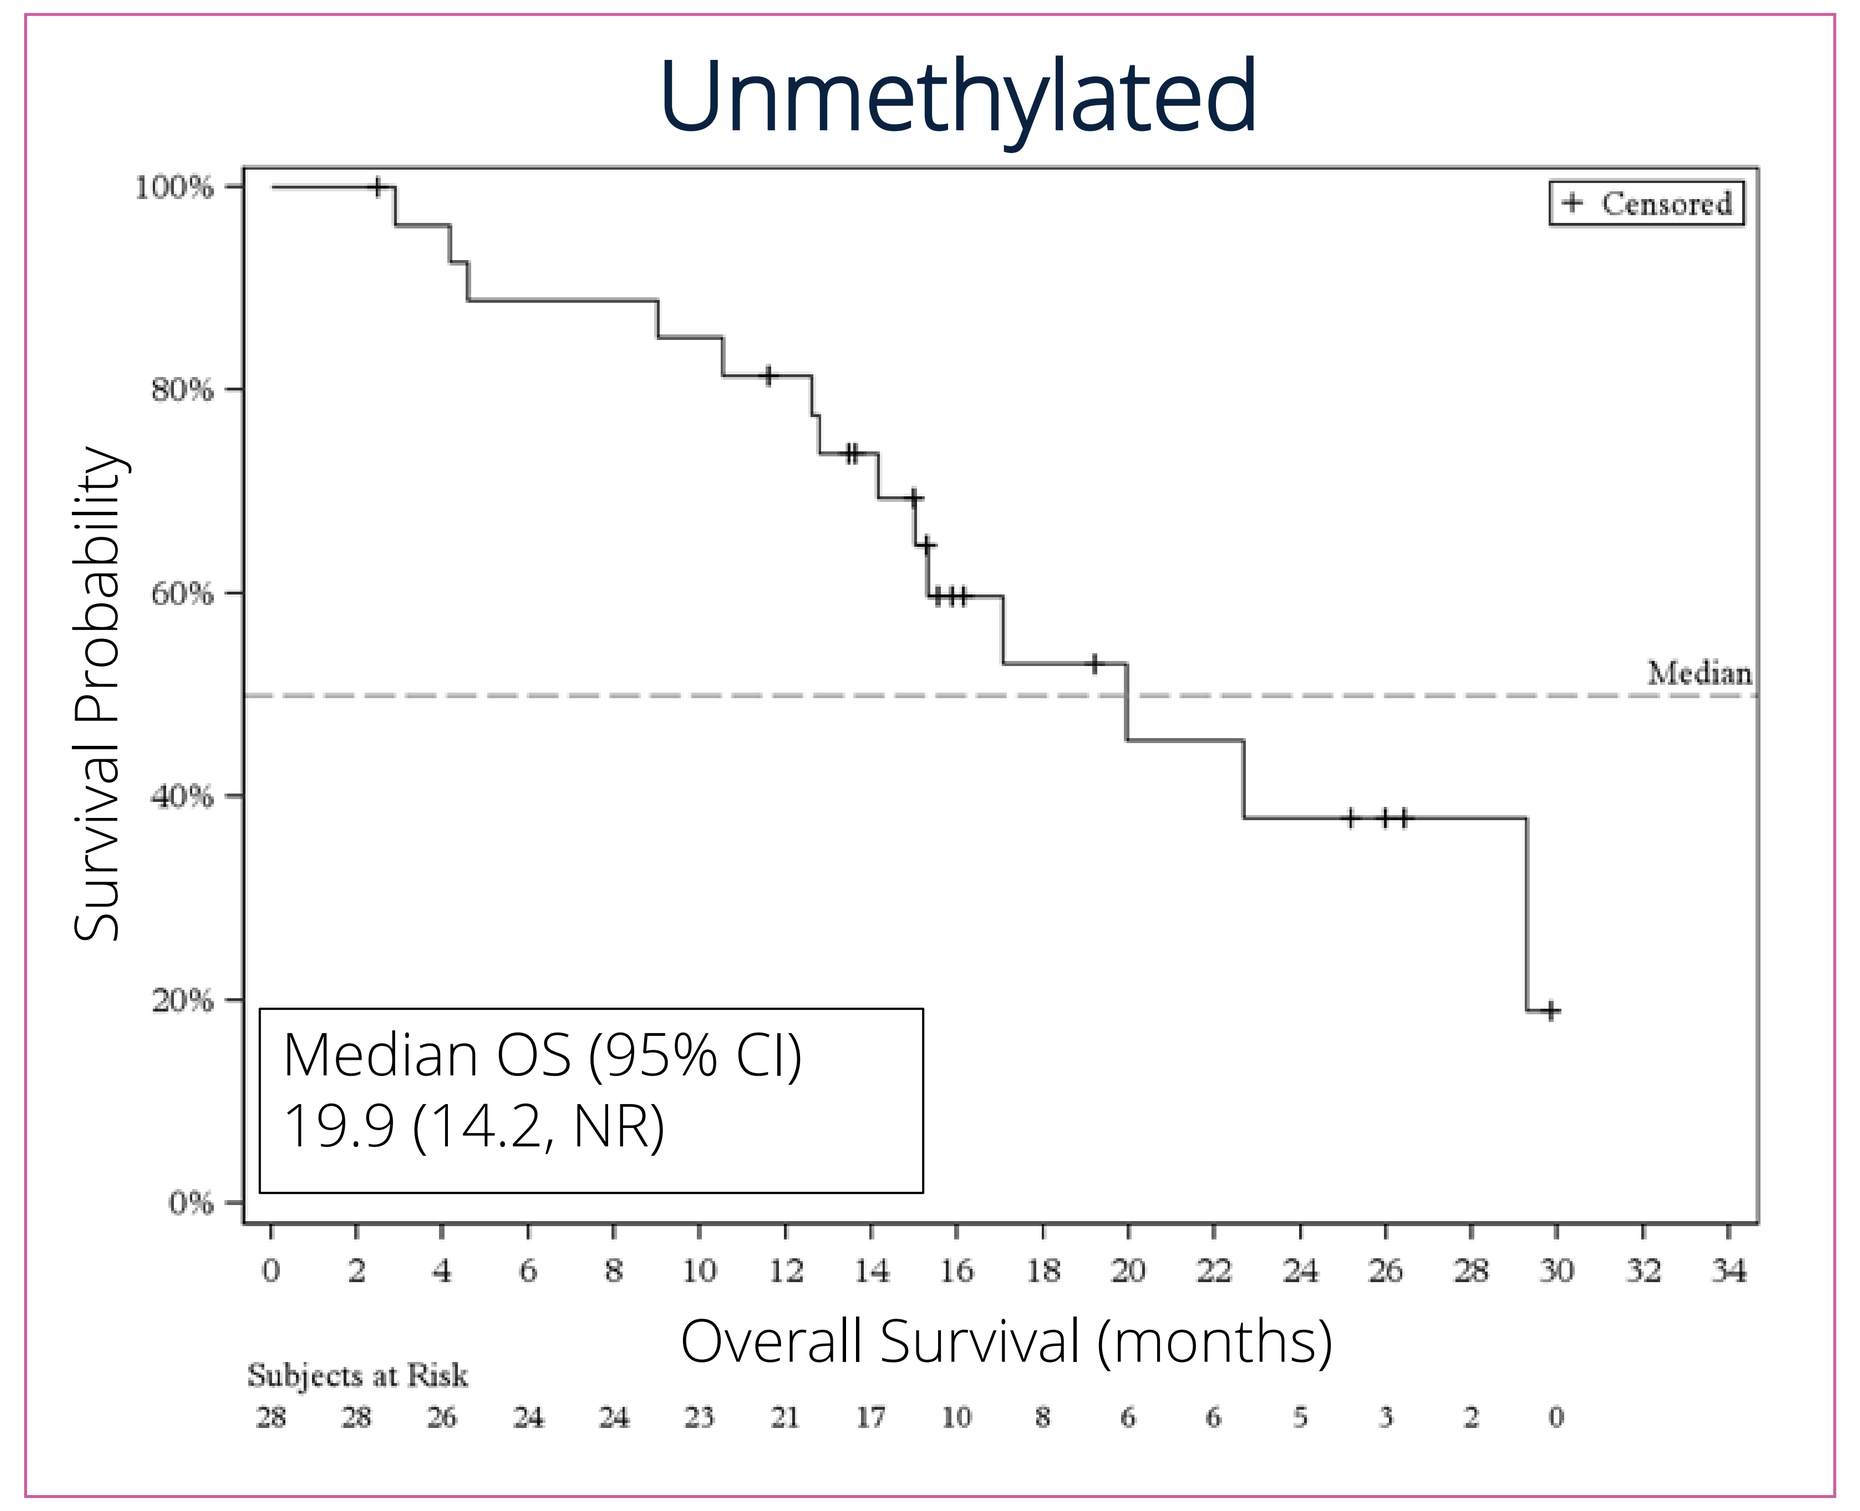
Unmethylated
+ Censored
Median
Survival Probability
Median OS (95% CI)
19.9 (14.2, NR)
Overall Survival (months)
Subjects at Risk
28 28 26 24 24 23 21 17 10 8 6 6 5 3 2 0

PP= Per Protocol (subjects completed at least 2 weeks BPM31510 and started RT and TMZ), CI = Confidence Interval, OS = Overall Survival, NR = Not Reached; OS is the time from first date of BPM31510 exposure to death

Conclusions

- BPM31510 was well-tolerated and does not exacerbate toxicity profile of RT +TMZ in a front-line setting.
- Median OS of ~20 months observed in **unmethylated** patients is a clinically meaningful improvement in OS compared to historical cohorts shown below.

	MGMT - Unmethylated	
Study	N	Median OS (95% CI)
This Study	24	19.9 (14.2, NR)
Gilbert'13*	254	14.60 (13.20, 16.50)
Gilbert'14*	214	14.60 (13.60, 15.60)
Stupp'05*	60	12.70 (11.60, 14.40)
Stupp'17*	95	14.70 (12.80, 19.10)
Westphal'15*	32	15.50 (13.80, 24.00)

*Alnahhas, Iyad et al. *Neuro-oncology advances* vol. 2,1 vdaa082. 30 Oct. 2020, doi:10.1093/noajnl/vdaa082

- BPM31510IV-11 (NCT04752813) has completed recruiting.
- Planning for a future registrational trial is underway.

Corresponding author email address Dr. Seema Nagpal, snagpal@stanford.edu
Dr. Nagpal and Dr. Stocksdale receive institutional research support from BPGbio; Study sponsored by BPGbio
The safety and efficacy of BPM31510 has not been determined yet by the FDA.